A PHASE 1/2 TRIAL OF SCIB1 IMMUNOBODY IN MELANOMA PATIENTS

PM Patel[1], LG Durrant[1,2], C Ottensmeier[3], C Mulatero[4], P Lorigan[5], R Plummer[6], M Cunnell[1], R Metheringham[2], V Brentville[2], L Machado[2], I Daniels[2] and D Hannaman[7]

[1]University of Nottingham, Nottingham UK, [2] Scancell Ltd, Nottingham UK, [3]Southampton University Hospital NHS Foundation Trust, Southampton UK, [4]St James University Hospital, Leeds UK, [5]The Christie Hospital NHS Foundation Trust, Manchester UK, [6]Northern Institute for Cancer Research, Newcastle Upon Tyne UK, [7]Ichor Medical Systems Inc., San Diego CA

PROTOCOL

ImmunoBody® is a DNA vaccine encoding a human IgG1 antibody with melanoma-specific T-cell epitopes grafted into its CDR regions. SCIB1 has a single epitope grafted from the TRP2 antigen and three grafted from gp100. This vaccine targets dendritic cells *in vivo* and stimulates high avidity T-cells which result in the elimination of established tumors in pre-clinical models. A clinical trial was conducted to evaluate the safety of SCIB1 and its ability to induce a cellular immune response.

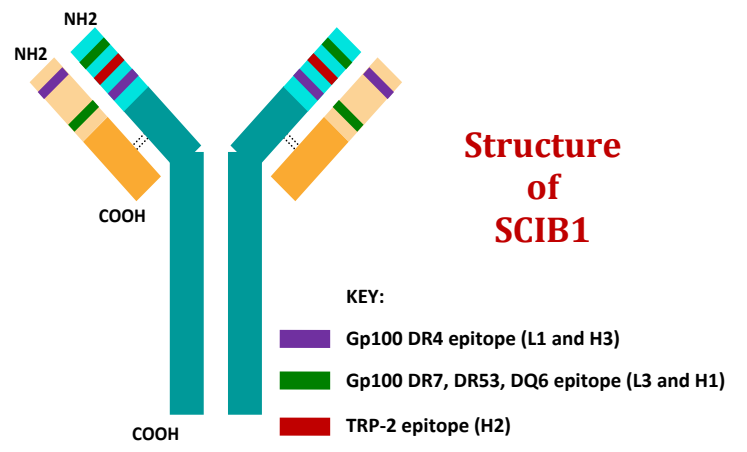
NH2
NH2
COOH
COOH
KEY:
Gp100 DR4 epitope (L1 and H3)
Gp100 DR7, DR53, DQ6 epitope (L3 and H1)
TRP-2 epitope (H2)

Structure of SCIB1

Primary Objective:
To demonstrate safety and tolerability

Secondary Objectives:
To demonstrate cellular immune response
To assess tumor response

Study Design

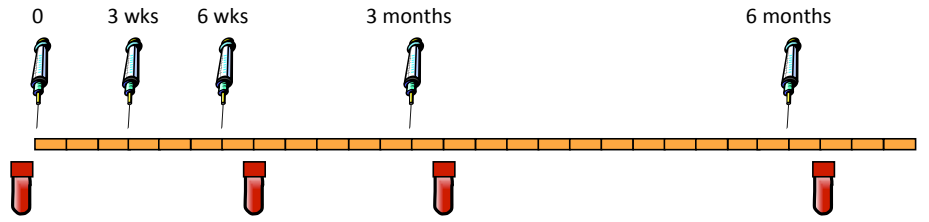
0
3 wks
6 wks
3 months
6 months

- Intramuscular injection with electroporation
- Initially administered at 3-weekly intervals for three injections, then given after 3 and 6 months
- Option for continuation treatment every 3-6 months

PART ONE - Cohorts 1, 2, 3
- Nine patients with stage III/IV metastatic disease
- Three subjects per cohort (0.4, 2.0 or 4.0 mg dose)
- Progression if safety demonstrated at previous dose
- 2 mg patients allowed to progress to 4 mg

PART ONE - Cohort 4
- Five patients with stage IV metastatic disease
- 8 mg dose
- One patient on continuation therapy, one due to start

PART TWO
- 14 stage III/IV patients recruited (4 mg dose)
- Tumors resected before treatment
- Six patients on continuation therapy

SCIB1 SAFETY PROFILE

- No dose limiting toxicities were observed
- No CTC grade 4/5 toxicities
- Most common adverse events were injection site reactions (pain, tenderness, bruising, erythema and swelling)

CLINICAL RESPONSES

- Part 1 stage III/IV patients receiving >3 doses of 2 mg/4 mg SCIB1:
 - 4/6 patients still alive with a median survival time of 30 months from study entry
 - 1-year survival = 100% and 2-year survival = 67%
- Part 1 (8 mg dose) stage III/IV patients with tumor present:
 - All 5 patients remain alive
 - Median survival time is 10 months from study entry
- Part 2 (4 mg dose) stage III/IV patients with resected tumors:
 - Median overall survival has not yet been reached; survival at 1-year =100%
 - All 14 patients remain alive
 - Only three patients have evidence of disease progression after 4, 14 and 18 months on study
 - Median survival time is 21 months from study entry, 26 months from diagnosis of metastatic disease
- Evidence of a clinical response seen in 5/14 patients in Part 1:
 - One patient (04-16) receiving 4 mg doses had multiple tumor lesions which all decreased in size or disappeared except for one lesion which was resected – immunohistochemistry demonstrated loss of gp100 and strong expression of PD.L1
 - One patient (04-28) receiving 8 mg doses has shown a partial response of all lesions and is scheduled to start continuation treatment
 - One patient (receiving 2 mg/4 mg doses) has been disease-free for 33 months having previously been treated every year for 5 years with recurrent skin and lymph node disease
 - One patient (receiving 4 mg doses) remained disease-free for 18 months having previously required the resection of multiple metastases
 - A further patient with lung and breast tumors at study entry remained stable for 6 months after receiving 8 mg doses of SCIB1 and is receiving continuation treatment

CT lung sections from Part 1 patients demonstrate a reduction in lesion size

Patient 04-16: (i) Pre-treatment, (ii) 9 months
Patient 04-28: (i) Pre-treatment, (ii) 6 months

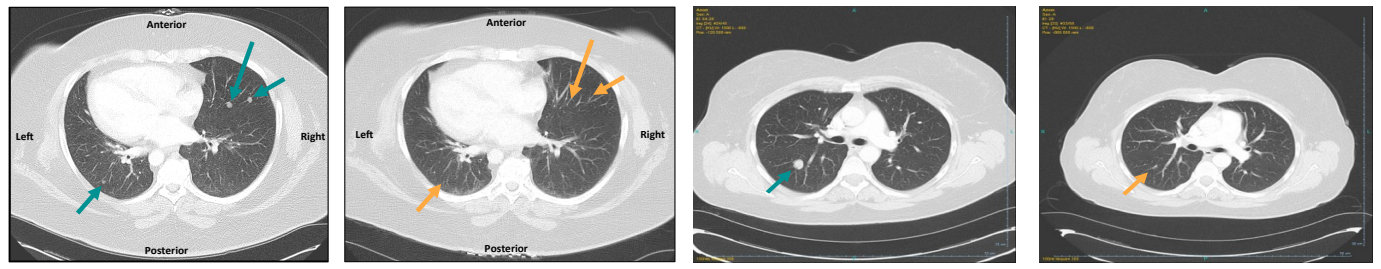
Anterior
Left
Right
Posterior
Anterior
Left
Right
Posterior

SUMMARY OF CLINICAL RESPONSES

Part 1 patients

Pt	Dose (mg)	No doses	Survival since study entry	Survival since diagnosis of mets	Treatment post-SCIB1	Status	Disease Status
01-01	0.4	4	7	34	-	-	PD
02-01	0.4	4	16	67	Ipilimumab	-	PD
02-04	0.4	3	7	20	-	-	PD
01-16	2/4	3 + 2	14	19	-	-	PD
01-19	2/4	4 + 1	38	72	Verumafenib (non-responder)	Alive	PD
01-24	2/4	3 + 2	34	55	-	Alive	DF
04-03	4	5	32	34	Ipilimumab	Alive	PD
05-05	4	5	13	16	-	-	PD
04-16	4	5	27	68	No	Alive	MR
Survival (median, months) Cohorts 2 and 3			30	48			
01-46	8	5	8	62	-	Alive	PD
04-27	4/8	2 + 3	10	78	-	Alive	SD
04-28	8	5	8	12	-	Alive	PR
05-26	8	3	11	22	Ipilumumab	Alive	PD
05-27	8	4	11	33	-	Alive	PD
Survival (median, months) Cohorts 4			10	33			

Part 2 patients

Pt	Dose (mg)	No doses	Survival since study entry	Survival since diagnosis of mets	Treatment post-SCIB1	Status	Disease Status
01-32	4	5	22	25	-	Alive	DF
01-34	4	5	24	28	-	Alive	DF
01-37	4	5	23	141	-	Alive	Recurrence
02-21	4	5	22	26	-	Alive	DF
02-33	4	5	17	25	-	Alive	DF
04-22	2	3	19	26	-	Alive	DF
05-08	4	5	24	28	-	Alive	Recurrence
05-09	4/2	4 + 1	23	26	Surgery	Alive	Recurrence
05-11	4	5	18	27	-	Alive	DF
05-13	4	5	22	28	-	Alive	DF
05-18	4	5	15	24	-	Alive	DF
05-19	4	5	18	24	-	Alive	DF
05-21	4	5	19	42	-	Alive	DF
05-24	4	5	18	19	-	Alive	DF
Survival (median, months) Part 2			21	26			

PD = progressive disease
PR = partial response
DF = disease free
SD = stable disease
MR = mixed response
Data cut-off 16 April 2014

Overall survival and Progression-free survival in Stage III/IV melanoma patients

Part 1 (n=16): Overall survival
Part 2: (n=14) Progression-free survival

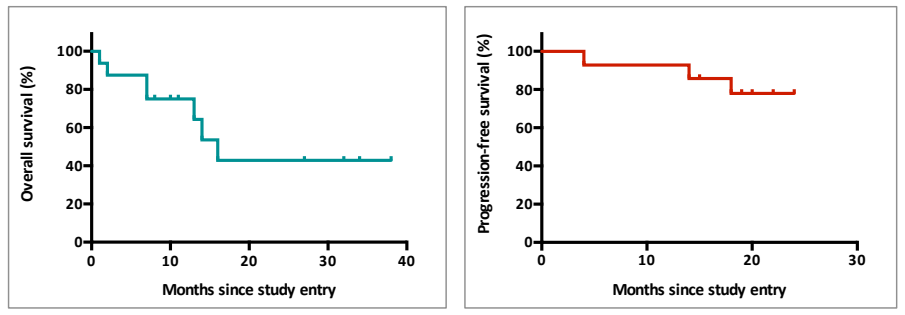
Overall survival (%)
Months since study entry
Progression-free survival (%)
Months since study entry

CONCLUSIONS

- SCIB1 is safe and well tolerated in late stage melanoma patients
- 24 of 28 evaluable patients developed melanoma-specific immune responses following repeat dosing with SCIB1
- Detection of two objective clinical responses in conjunction with evidence of disease control and overall survival times is highly encouraging
- Stimulation of a potent *de novo* immune response by SCIB1 may synergise with anti-PD1 checkpoint inhibitors to increase response rates

IMMUNE RESPONSES

- **Part 1**
 - One patient in the 0.4 mg cohort, all three patients in the 2 mg cohort and two patients in the 4 mg cohort mounted an immune response to the vaccine antigens
 - Four out of five patients in the 8 mg cohort made an IFNγ Elispot response after T cell expansion *in vitro*
 - Elispot responses to the 8 mg dose were up to 10-fold higher than those seen in the 4 mg groups, with frequencies exceeding 2% of total blood lymphocytes
- **Part 2**
 - All 14 patients generated an immune response
 - 13 patients responded in the proliferation assay; 13 patients responded in the Elispot assay; 12 patients responded in both assays

Proliferation responses

Melanoma-specific responses in PBMCs were assayed on days 7 and 11 by the incorporation of tritiated thymidine. Data presented from patient 05-13 (Part 2)

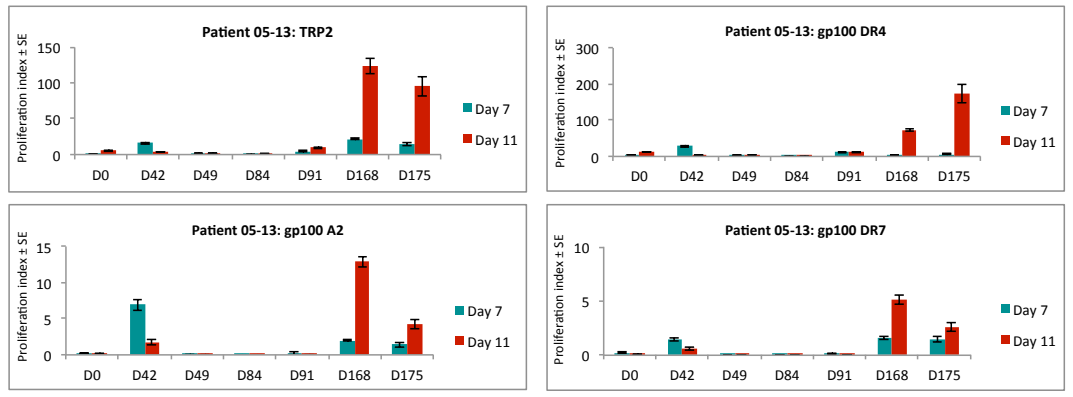
Patient 05-13: TRP2
Patient 05-13: gp100 DR4
Patient 05-13: gp100 A2
Patient 05-13: gp100 DR7
Proliferation index ± SE
D0 D42 D49 D84 D91 D168 D175
Day 7
Day 11

Elispot responses

The generation of IFN gamma by cultured PBMCs was assayed on day 9 and day 16 by Elispot. Data presented from patient 04-28 (Part 1, cohort 4)

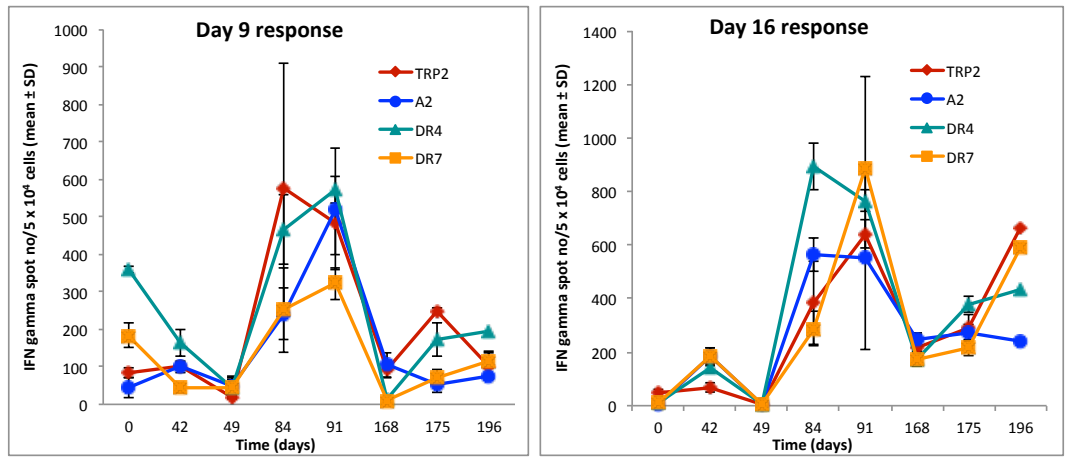
Day 9 response
Day 16 response
IFN gamma spot no/5 x 10⁴ cells (mean ± SD)
Time (days)
0 42 49 84 91 168 175 196
TRP2
A2
DR4
DR7

OVERALL IMMUNE RESPONSE SUMMARY

	No of Patients	No of Immune Responders	Number of Epitopes recognised by individual patients: 1	2	3	4
Part 1	**14**	**10**	**3**	**1**	**1**	**5**
Cohort 1	3	1	1	-	-	-
Cohort 2	3	3	1	-	3	1
Cohort 3	3	2	1	-	-	1
Cohort 4	5	4	-	1	-	3
Part 2	**14**	**14**	**-**	**1**	**3**	**10**
Total	**28**	**24**	**3**	**2**	**4**	**15**